Tolerability and Infusion Duration of Pegunigalsidase Alfa in Patients with Fabry Disease: Data from 5 Completed Clinical Trials

John Bernat[1], Myrl Holida[1], Ozlem Goker-Alpan[2], Ankit Mehta[3], Nicola Longo[4], Eric Wallace[5], Khan Nedd[6], Mirjam Langeveld[7], William Wilcox[8], Derralynn Hughes[9], Rossana Rocco[10], Irene Koulinska[11], Einat Almon[12], Sari Alon[12], Raul Chertkoff[12], Pilar Giraldo[13], Antonio Pisani[14], Ales Linhart[15]

[1]University of Iowa Hospital and Clinics, Iowa City, IA, USA; [2]LRDRT Center, Fairfax, VA, USA; [3]Baylor University Medical Center, Dallas, TX, USA; [4]University of Utah, Salt Lake City, UT, USA; [5]University of Alabama at Birmingham, Birmingham, AL, USA; [6]Infusion Associates, Grand Rapids, MI, USA; [7]Academisch Medisch Centrum Universiteit van Amsterdam, Netherlands; [8]Emory University School of Medicine, Atlanta, GA, USA; [9]LSDU, Royal Free London NHS Foundation Trust, and University College London, England, UK; [10]Chiesi Farmaceutici S.p.A., Parma, Italy; [11]Chiesi USA, Inc., Boston, MA, USA; [12]Protalix Biotherapeutics, Carmiel, Israel; [13]Hospital de Dia Quiron, Zaragoza, Spain; [14]University of Naples Federico II, Naples, Italy; [15]Charles University, Prague, Czech Republic

Presenting person: Annarita Armaroli,* Chiesi Italy S.p.a, Parma Italy

Introduction

- Fabry disease is a rare X-linked genetic disorder caused by deficiency in lysosomal enzyme alpha-galactosidase A (α-Gal A) activity[1]
- This deficiency results in accumulation of glycosphingolipids, mainly globotriaosylceramide (Gb3), which can lead to multisystemic disease and early death[1]
- Pegunigalsidase alfa is a novel PEGylated α-Gal A enzyme replacement therapy (ERT) in development for the treatment of patients with Fabry disease that was designed to offer prolonged half-life and lower immunogenicity with potential tolerability benefits[1]
- These features of pegunigalsidase alfa may reduce immunogenicity that would result in a lower incidence of infusion-related reactions (IRR) and demonstrate improved overall tolerability, compared with other ERTs on the market,[2] when infused at the same dose (1.0 mg/kg)
- Theoretically, patients previously treated with other ERTs can potentially experience IRRs when switched to pegunigalsidase alfa because of differences in dose or differences in immunogenicity between therapies

Objective

- To characterize the tolerability profile of pegunigalsidase alfa using data from 5 clinical trials that had been completed by 2021, with a focus on the incidence of IRRs and change in infusion duration over time

Methods

- Patients were included from 5 pegunigalsidase alfa clinical trials **(Table 1)**
- IRRs were defined as possibly, probably, or definitely related treatment-emergent adverse events (TEAEs) occurring during the infusion or within 2 hours post infusion (excluding those TEAEs defined as injection-site reactions)
- The following outcomes are described:
 – Incidence of IRRs
 – Mean duration of infusions at beginning and end of study
 – Number of patients achieving the minimum duration of infusion allowed per protocol
 – Time to achieve the minimum per-protocol infusion duration

Table 1. Study Design Summaries and Patient Baseline Characteristics

Study	Clinical trials NCT #	Study design	Previous ERT-treatment status	Study duration, months	Pegunigalsidase alfa dose regimen, mg/kg, and frequency	Men, n	Women, n	Overall, N	Mean age, years (SD)
F01/F02	NCT01678898 NCT01769001	Open-label, dose-ranging studies in treatment-naïve patients	None	F01: 3 F02: 9	0.2 EOW 1.0 EOW 2.0 EOW	4 6 1	2 2 3	6 8 4	30.0 (10.8) 33.5 (11.7) 40.0 (16.5)
Extension F03	NCT01981720	Open-label extension of F01/F02	Yes: pegunigalsidase alfa for up to 60 months	≤ 60	1.0 EOW	8	7	15	33.4 (12.5)
BRIDGE	NCT03018730	Open-label, single-arm, switchover from agalsidase alfa	Yes: agalsidase alfa for 2+ years	12	1.0 EOW	15	7	22	34.8 (11.9)
BRIGHT	NCT03180840	Open-label, single-arm, switchover from other approved ERTs	Yes: agalsidase beta or agalsidase alfa for 3+ years	12	2.0 Q4W	24	6	30	40.5 (11.3)

EOW, every other week; ERT, enzyme replacement therapy; Q4W, every 4 weeks; SD, standard deviation.

Results

Infusion Duration

- In the F01 and F02 studies, 18 treatment-naïve patients received pegunigalsidase alfa every other week (EOW):
 – Initial mean infusion durations were 4.0, 5.5, and 6.4 hours for pegunigalsidase alfa 0.2 mg/kg, 1.0 mg/kg, and 2.0 mg/kg, respectively (**Figure 1**)
 – Mean infusion durations were decreased at 12 months to 1.5, 3.3, and 3.1 hours for pegunigalsidase alfa 0.2 mg/kg, 1.0 mg/kg, and 2.0 mg/kg, respectively.
- In the F03 extension, all patients (n = 15) received pegunigalsidase alfa 1.0 mg/kg infusions EOW, and 13 achieved protocol-specified minimum 1.5-hour infusion durations between months 12 and 22.

- In the BRIDGE study, patients (n = 22) receiving pegunigalsidase alfa 1.0 mg/kg EOW had an initial mean infusion duration of 2.9 hours (standard deviation [SD] 0.9 h) (**Figure 2**):
 –2 Patients discontinued treatment owing to type 1 IgE mediated- hypersensitivity reactions that occurred within 2 hours of the first infusion.
 –All 20 patients who completed the study reached the minimum protocol-allowed infusion duration of 1.5 hours ± 10 minutes by 12 months ($p <$ 0.001 vs baseline).

- In the BRIGHT study, 30 patients initially received pegunigalsidase alfa 2.0 mg/kg every 4 weeks:
 – Mean infusion duration decreased from 4.8 hours (n = 30; SD 0.59 h) at baseline to 2.3 hours (n = 29; SD 0.7 h; $p < 0.001$) by month 12 (**Figure 3**)
 – At study completion, most patients had reached targeted infusion durations of:
 - 3 Hours for patients weighing > 100 kg (n = 5): mean of 2.97 hours (SD 0.24 h); 100% of patients reached target
 - 2 Hours for patients weighing ≤ 100 kg (n = 24): mean 2.1 hours (SD 0.13); 96% of patients reached target
 - 1 Patient discontinued because of a motor vehicle accident and 1 patient did not reach target infusion time

Figure 1. F01/F02/F03 Studies: Mean Infusion Duration Over Time of Pegunigalsidase Alfa EOW (n = 15)

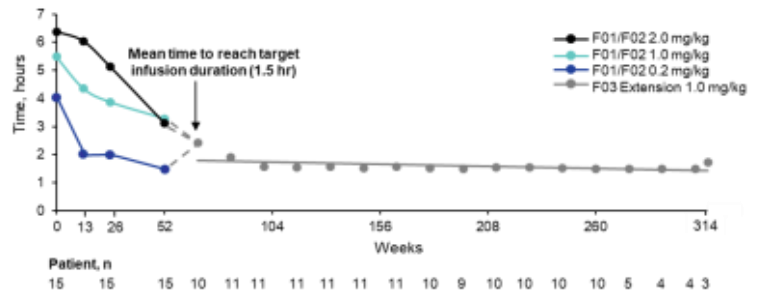
Time, hours
Mean time to reach target infusion duration (1.5 hr)
F01/F02 2.0 mg/kg
F01/F02 1.0 mg/kg
F01/F02 0.2 mg/kg
F03 Extension 1.0 mg/kg
Weeks
Patient, n
15 15 15 10 11 11 11 11 11 11 10 9 10 10 10 10 5 4 4 3

F03 extension data represent the mean infusion values for 15 patients receiving pegunigalsidase alfa 1.0 mg/kg over 12 to 72 months. The grey line represents the best fit of infusion duration values from F03

Figure 2. BRIDGE Study: Mean Infusion Duration Over Time of Pegunigalsidase Alfa 1.0 mg/kg EOW (n = 20)

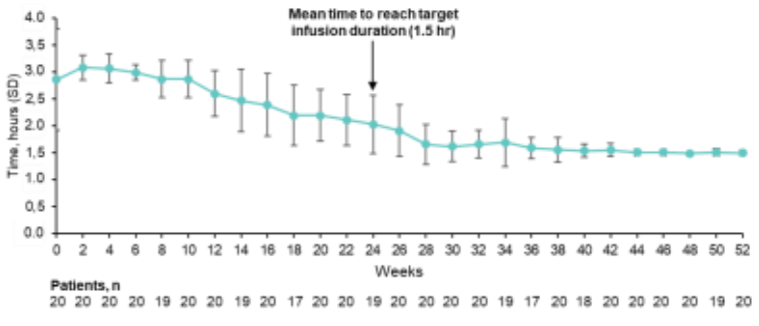
Time, hours (SD)
Mean time to reach target infusion duration (1.5 hr)
Weeks
Patients, n
20 20 20 20 19 20 20 19 20 17 20 20 19 20 20 20 20 19 17 20 18 20 20 20 20 19 20

n = 20 at 12 months; $p < 0.001$ from baseline to 12 months.
EOW, every other week; SD, standard deviation.

Figure 3. BRIGHT Study: Mean Infusion Duration Over Time of Pegunigalsidase Alfa 2.0 mg/kg Q4W (n = 29)

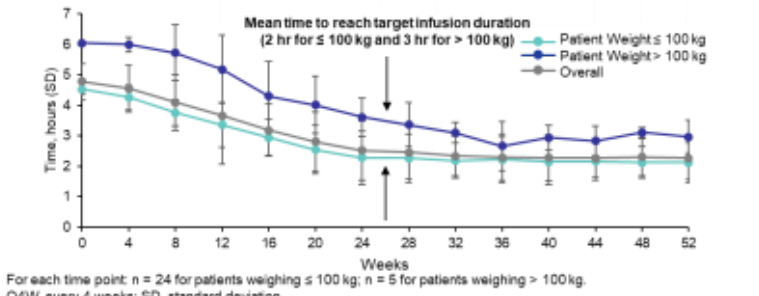
Time, hours (SD)
Mean time to reach target infusion duration (2 hr for ≤ 100 kg and 3 hr for > 100 kg)
Patient Weight ≤ 100 kg
Patient Weight > 100 kg
Overall
Weeks

For each time point: n = 24 for patients weighing ≤ 100 kg; n = 5 for patients weighing > 100 kg.
Q4W, every 4 weeks; SD, standard deviation.

Safety

- F01/F02 Study:
 – 5 Patients (28% [n = 3] women) experienced 24 IRRs (**Table 2**)
 - With 1 exception, all were of mild or moderate severity
 - There was 1 serious adverse event (SAE) of an IgE mediated-hypersensitivity reaction (bronchospasm) during the first infusion that resulted in a discontinuation per study protocol

Table 2. F01/F02 Studies: Infusion-Related Reactions

PA dose, mg/kg	Sex	IRR Severity classification	Visit number	IRR Description	IRRs, n
0.2	Female	Mild or moderate	1, 3	Chest tightening / discomfort	5
			1	Sneezing	
			3	Nausea	
			27	Sinus drainage / paranasal sinus hypersecretion	
1.0	Female	Mild or moderate	2, 15	Hypotension	5
			3	Lightheadedness / dizziness	
			9	Short of breath / dyspnea	
			13	Maculo-papular erythematous rash	
	Male	Mild or moderate	6, 8	Infusion reaction / IRR	6
			15	Pain at left chest / chest pain	
			17	Itching / pruritus	
			20	Nausea	
			20	Dizziness	
	Male[a]	Serious adverse event (Severe)	1	Bronchospasm	1
2.0	Female	Mild or moderate	1–7	Abdominal cramping / abdominal pain	7
Total, n					**24**
Total IRR incidence, %					**28**

[a]Patient discontinued.
IRR, infusion-related reaction; PA, pegunigalsidase alfa.

- F03 Extension Study:
 – 1 IRR of mild peripheral swelling was reported in 1 male patient (12.5%)
 - Among patients who had IRRs (n = 6), 3 (50%) were IgG ADA positive[a]
 - Among patients without IRRs (n = 9), 3 (33.3%) were IgG ADA positive[a]

- BRIDGE Study:
 – 9 IRRs were reported in 5 patients (22.7%; all men) (**Table 3**)
 - Of patients who had IRRs (n = 5), 2 (40%) were IgG ADA positive[a]
 - Of patients without IRRs (n = 17), 5 (29.4%) were IgG ADA positive[a]
 - Most IRRs (7) were nonserious and of mild severity
 - 2 IRRs experienced by 2 male patients were severe and serious
 - Both were type 1 IgE mediated-hypersensitivity reactions and led to discontinuation
 - 1 Patient was positive for immunoglobulin E antidrug antibodies at baseline
- BRIGHT Study:
 – 27 IRRs were reported in 5 patients (17%; all men) (**Table 4**)
 - Of patients who had IRRs (n = 5), 4 (80%) were IgG ADA positive[a]
 - Of patients without IRRs (n = 25), 6 (24%) were IgG ADA positive[a]
 - 17 IRRs were mild and 10 were of moderate severity
 - All patients completed the study

[a]IgG ADA positive at any time including baseline or any follow up visits.

Table 3. BRIDGE Study: Infusion-Related Reactions

PA dose, mg/kg	Sex	IRR Severity classification	Visit number	IRR Description	IRRs, n
1.0	Male	Mild	4	Itching/Rash pruritus	5
			5	Erythema	
			9	Pruritus	
			10, 11	IRR	
	Male[a]	Serious adverse event (severe)	1	Type 1 IgE mediated-hypersensitivity reaction	1
	Male[a]	Serious adverse event (severe)	1	Type 1 IgE mediated-hypersensitivity reaction	1
	Male	Mild	20	Nasal congestion	1
	Male	Mild	19	Dizziness	1
Total, n					**9**
Total IRR incidence, %					**23**

Table 4. BRIGHT Study: Infusion-Related Reactions

PA dose, mg/kg	IRR Severity classification	IRR Description	Patients (n = 5; all men)	IRRs, n
2.0	Mild	Infusion-related reaction	1	11
		Nausea	1	1
		Asthenia	1	1
		Pain	1	1
		Chest discomfort	1	1
		Paresthesia	1	1
		Tremor	1	1
			Total	17
	Moderate	Infusion-related reaction	3	4
		Pain	1	1
		Hypersensitivity	1	1
		Vomiting	1	1
		Myalgia	1	1
		Pain in extremity	1	1
		Headache	1	1
			Total	10
Overall total, n				**27**
Total IRR incidence, %				**17**

IRR, infusion-related reaction; PA, pegunigalsidase alfa.

Conclusions

- This analysis of data from studies completed by 2021 supports the favorable tolerability profile of pegunigalsidase alfa in patients with Fabry disease
- Mean infusion durations by study ends were reduced and/or met target rates
- IRR incidence in pegunigalsidase alfa trials completed by 2021 ranged from 17% in BRIGHT to 28% in F01/02, which compares favorably to available ERTs administered at the same dose (55%–67%)[2,3]
- The percentage of patients who were IgG ADA positive among those with IRRs were higher than among those without IRRs, especially in BRIGHT study
- Most (57/60) IRRs reported during pegunigalsidase alfa treatment were of mild or moderate severity
- Overall, SAEs were reported in 3 of the 60 IRRs: one bronchospasm and two Type I hypersensitivity reactions, all were IgE mediated
- Additional data from the pegunigalsidase alfa clinical program may further characterize its potential to offer both tolerability and improved quality-of-life benefits for patients with Fabry disease

References
1. Schiffmann R et al. *J Inherit Metab Dis*. 2019;42(3):534-544. 2. Fabrazyme® (agalsidase beta injection, for intravenous use). Genzyme Corporation; 2021. Accessed April 26, 2022. https://products.sanofi.us/Fabrazyme/Fabrazyme.pdf 3. Fabrazyme® (agalsidase beta injection powder). Genzyme Europe B.V. Accessed April 26, 2002. https://www.ema.europa.eu/en/documents/product-information/fabrazyme-epar-product-information_en.pdf

Acknowledgments: This study was sponsored by Protalix. Medical writing support was provided by Ebony Lai Hing, PhD, of Oxford PharmaGenesis Inc., Newtown, PA, USA, and was funded by Chiesi USA, Inc.

Disclosures: **JB** receives research support from Avrobio, BioMarin Pharmaceutical, Idorsia Pharmaceuticals, Pfizer, Protalix Biotherapeutics, Sanofi, and Takeda; and has participated in advisory boards for Sanofi, Takeda, and Travere Therapeutics. **MH** received speaker-related fees from Protalix and has been, or is currently, involved in clinical trials with Sanofi, Sangamo, Avrobio, Protalix, and Idorsia (no direct funding is received for these trials as they are institution directed). **OG-A** has conducted contracted research, received consulting fees, and/or served on advisory boards with Amicus, Freeline, Genentech, Protalix, Sangamo, Sanofi, Takeda, 4DMT, and Avrobio. **AM** reports non-financial support from Chiesi Pharmaceuticals, during the conduct of the study; personal fees from AstraZeneca, personal fees from Bayer Pharmaceuticals, personal fees from Janssen Pharmaceuticals, outside the submitted work **NL** receives research support from and has participated in advisory boards for Amicus, Astellas, Avrobio, BioMarin Pharmaceutical, Homology, Horizon, Moderna, Pfizer, Protalix Biotherapeutics, PTC Biotherapeutics, Reneo, Sanofi, Takeda, and Ultragenyx (no direct funding is received as they are institution directed). **EW** has consulting and/or grants with Sanofi, Protalix, Chiesi, Idorsia, 4DMT, Amicus, and Natera. **KN** has nothing to disclose. **ML** is involved in premarketing studies with Sanofi, Protalix, and Idorsia. Financial arrangements were made through AMC Research BV. No fees, travel support or grants were obtained from Pharmaceutical Industry. **WW** Consultant (advisory boards): Amicus, Chiesi, Sanofi, Spark, Takeda, UniQure. Clinical trials and registries: Amicus, Chiesi, 4D Molecular Therapeutics, Protalix, Sanofi-Genzyme, Sangamo, and Takeda. Research grants: Amicus and Takeda. **DH** has received honoraria for speaking and consulting fees for advisory boards from Protalix, Takeda, Sanofi, Freeline, and Sangamo, administered through University College London consultants, and used in part to support research in lysosomal storage diseases. **RR** is a full-time employee of Chiesi Farmaceutici S.p.A., Parma, Italy. **IK** is a full-time employee of Chiesi, USA. EBA, SA and RC are full-time employees of Protalix Biotherapeutics. PG has been involved in premarketing studies with Genzyme, Protalix, and Idorsia, and has received grants from Sanofi and Takeda; monies received for these activities have been deposited into the Spanish Foundation for the Study and Treatment of Gaucher Disease (FEETEG) to contribute to the development of research in lysosomal storage disorders. **AP** travel expenses and grant from Takeda, Sanofi, Amicus, and Chiesi. **AL** has received consultancy and speaker's honoraria from Amicus Therapeutics, Sanofi, Avrobio, and Takeda. **GD** received honoraria, travel support, and accommodation from Amicus Therapeutics, Greenovation Biotech GmbH, Protalix, Sanofi, and Takeda. *Presenting person **AA** is a Full employee of Chiesi Italy.